Pharmacokinetics and Safety of Single, Multiple, and Loading Doses of CEM-102 in Healthy Subjects

Poster # A1-1931

JG Still [1], K Clark[1], TP Degenhardt [1], D Scott [1], P Fernandes [1], MJ Gutierrez [2],

[1] Cempra Pharmaceuticals, Inc., Chapel Hill, NC, [2] Comprehensive Phase One, Miramar, FL

J. Gordon Still, MD, PhD
6340 Quadrangle Dr., Suite 100
Chapel Hill, NC 27517
(919) 467-1716
gstill@cempra.com

Revised Abstract

Background: CEM-102 is an antibiotic of the fusidane class under development for treatment of acute bacterial skin structure infections. PK and safety of escalating single (SD), multiple (MD), and loading dose (LD) regimens of oral CEM-102 were evaluated.

Methods: This was a randomized, double-blind, placebo-controlled, dose escalation study. CEM-102 doses (550, 1100, 1650, 2200 mg) were administered as SD, then BID (up to 1650 mg) for 5.5 days to 4 groups of healthy subjects in the fasting state. In addition, 2 LD regimens (1100 or 1650 mg BID on Day1 followed by 550 or 825 mg BID for 6.5 days) were evaluated. In each group 6 subjects received CEM-102 and 2 received placebo. Dose escalation occurred after safety of the previous dose was determined. Physical examinations, vital signs, ECGs, clinical laboratory tests, and adverse events (AEs) were monitored. Blood for assay of CEM-102 concentrations and PK analysis was collected pre-dose and at specified intervals after each dose.

Results: Mean C_{max} and AUC_{0-t} ranged from 33.4 µg/mL and 242 µg•h/mL to 128 µg/mL and 1690 µg•h/mL for SD of 550 to 2200 mg and from 130 µg/mL and 1150 µg•h/mL to 324 µg/mL and 3290 µg•h/mL for MD of 550 to 1650 mg. LD regimens resulted in trough concentrations on Days 2 and 8 of 73.8 and 101 µg/mL (1100/550 mg regimen) and 146 and 204 µg/mL (1650/825 mg regimen). All doses appeared to be safe. SD up to 1650 mg, MD up to 1100 mg BID, and both LD regimens were well-tolerated. Nausea and vomiting occurred at SD 2200 mg (1 subject) and MD 1650 mg (4 subjects), therefore MD above 1650 mg were not administered. No other clinically meaningful AEs were seen. There were no clinically significant changes in monitored safety parameters.

Conclusion: CEM-102 was safe at all doses. Plasma exposure was higher after MD compared to SD, indicating accumulation with MD. LD regimens were well-tolerated and produced plasma concentrations that approached steady-state at 24 hours.

Introduction

CEM-102 (Figure 1) is sodium fusidate, an antibiotic of the fusidane class that is under development by Cempra Pharmaceuticals for treatment of patients with acute bacterial skin structure infections. Systemic sodium fusidate has been used widely for many years in the UK and other parts of Western Europe, Australia, and Canada to treat infections caused by susceptible and methicillin-resistant staphylococci.

CEM-102 inhibits bacterial protein synthesis through inhibition of elongation factor G, a mechanism of action different than that of any anti-infective agent currently approved in the US. CEM-102 has shown excellent microbiological activity against *Staphylococcus aureus,* including methicillin resistant *S. aureus* (MRSA) (MIC_{90} 0.12 µg/mL). Coagulase-negative staphylococci (CoNS) with or without oxacillin-resistance (MIC_{90} 0.12 µg/mL) were all highly susceptible to CEM-102. The MIC range of CEM-102 against more than 100 isolates of Group A streptococci was 2 to 8 µg/mL. In experimental murine infection models, CEM-102 showed protective effects against systemic infection with *S. aureus* and was effective in reducing bacterial density in systemic and soft tissue infections.

Metabolite stability and reaction phenotyping studies have shown that hepatic metabolism is a likely route of clearance for CEM-102 and that CYP3A4 is the principal enzyme that metabolizes CEM-102. Animal toxicology studies have shown limited toxicological effects of CEM-102, primarily minor gastrointestinal (GI) tract and adrenal gland changes. Pulmonary and cardiovascular safety pharmacology studies with CEM-102 in animal models found no evidence of adverse effects.

In a previous Phase 1 study in healthy subjects, CEM-102 500 mg single doses were safe and well tolerated. Mean C_{max} was 27.4 µg/mL and $t_{1/2}$ was 11.8 hours. Food appeared to decrease the C_{max} of CEM-102 by approximately 23%, although total exposure, time to peak plasma concentration and $t_{1/2}$ were comparable when a CEM-102 500 mg dose was administered under fed or fasting conditions.

In a second study, CEM-102 500 mg administered orally TID for 4.5 days for a total of 13 doses was safe and generally well tolerated. After the final dose on Day 5, mean CEM-102 concentrations rose rapidly and peaked at ~3 hours post-dose, then declined slowly with an apparent $t_{1/2}$ of 19 hours. Maximum plasma concentrations ranged between 87.6 and 245 µg/mL. Trough levels rose steadily from Day 1 to Day 5 attaining 105 µg/mL at trough on Day 5 and 120 µg/mL at 8 hours post-dose. The continued rise in mean trough concentrations following the last dose suggested that steady state was not yet reached by Day 5.

Optimal treatment of skin structure infection pathogens *S. aureus* and Group A streptococci may require attainment of steady-state plasma CEM-102 trough levels of 80 to 100 µg/mL early in the treatment course. The present study evaluated the PK, safety, and tolerability of single doses (SD) and multiple doses (MD) of CEM-102 and explored loading dose (LD) regimens aimed at achieving this profile in healthy adult subjects.

Figure 1. Chemical Structure of CEM-102

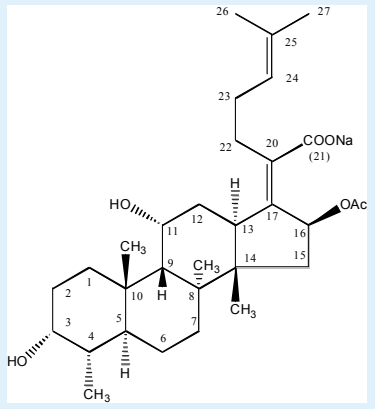

COONa
(21)
OAc
HO
CH3
H

Study Design and Methodology

Objectives

- To assess the PK, safety and tolerability of escalating SDs and MDs of oral CEM-102.
- To assess the PK, safety and tolerability of LDs of CEM-102 followed by lower Q12 hours (Q12h) maintenance doses.

Study Design and Selection Criteria

- Phase 1, randomized, double-blind, placebo-controlled, escalating-dose, SD, MD, and LD study in healthy adult male and female subjects.
- Enrolled: A total of 40 subjects were enrolled in 5 cohorts of 8 subjects.
- Key selection criteria:
 - ➢ Healthy males and females 18 to 55 years of age.
 - ➢ Body mass index (BMI) 18 to 30 kg/m^2; total body weight >60 kg.
 - ➢ (-) pregnancy test and appropriate contraceptives for females of childbearing potential.
 - ➢ Drug or alcohol abusers and tobacco users excluded.
 - ➢ Subjects with QTc >450 msec (470 msec for females) excluded.

Treatments

	Period 1	Period 2
Cohort 1:	550 mg X 1 → 7 day w/o* →	550 mg BID X 5.5 days
Cohort 2:	1100 mg X 1 → 7 day w/o →	1100 mg BID X 5.5 days
Cohort 3:	1650 mg X 1 → 7 day w/o →	1650 mg BID X 5.5 days
Cohort 4:	2200 mg X 1 → 7 day w/o →	1100 mg BID Day 1, then 550 mg BID X 6.5 days
Cohort 5:	No Period 1	1650 mg BID Day 1, then 825 mg BID X 6.5 days

- Within each dose group, 6 subjects received CEM-102 and 2 received placebo.
- Dose escalation proceeded only after the safety of the previous dose was determined.

* w/o = washout

Safety Assessments

- Safety was assessed by monitoring of adverse events (AEs), physical examinations, vital signs, and clinical laboratory tests pre- and post-dose. ECGs were obtained at baseline and anticipated time of peak plasma drug concentration.

Pharmacokinetic Assessments

- Blood samples for assay of CEM-102 concentrations and PK assessments were collected pre-dose and at frequent intervals up to 72 h post-dose after the single dose in Period 1 and the final dose in Period 2, and pre-dose and 3 h after each dose on Days 2-5 (Cohorts 1-3) and 2-7 (Cohorts 4-5) of Period 2.
- The following PK parameters were derived from the plasma concentration vs time curve after single doses and at steady state (as appropriate):
 - ➢ Maximum measured plasma concentration (C_{max})
 - ➢ Measured concentration prior to (C_{PRE}) and 12 hours following (C_{POST}) the 11th dose (Period 2) or 15th dose (Loading Dose Period 2)
 - ➢ Average concentration during a dosing interval at steady-state (C_{AVG})
 - ➢ Area under the concentration versus time curve (AUC_{0-t}, AUC_{0-tau}, and AUC_{∞})
 - ➢ The accumulation ratio (R_{ACC})
 - ➢ Time to peak concentration (t_{max})
 - ➢ Apparent terminal elimination half life ($t_{1/2}$)
 - ➢ Apparent first-order terminal elimination rate constant (Kel)
 - ➢ Apparent volume of distribution (V_d/F)
 - ➢ Apparent clearance (CL/F)

Statistical Methods and Data Analysis

- Sample size was based on clinical experience and judgment.
- No power calculations were performed.
- Data was summarized using descriptive statistics for continuous variables and frequencies and percentages for discrete variables.
- Graphs of mean plasma CEM-102 concentrations vs time were produced for each dose level and standard PK parameters calculated and summarized using descriptive statistics.

Results

Subjects

- Forty subjects (21 males and 19 females) were enrolled and 39 completed the study.
- Demographics were comparable in CEM-102 and placebo groups.

Safety and Tolerability

SDs	MDs and LDs:
• AEs were all mild in severity • No AEs seen at 1650 mg • GI AEs in 4 subjects at 2200 mg ➢ Nausea in 3 subjects ➢ Vomiting in 1 subject ➢ Abdominal distention in 1 subject • No clinically significant changes in labs, ECGs, or PEs • CEM-102 was well tolerated at single doses to 1650 mg	• AEs mostly mild in severity at all dose levels • GI symptoms (primarily mild nausea) in all MD groups ➢ 550 mg BID = 2 subjects ➢ 1100 mg BID = 4 subjects ➢ 1650 mg BID = 5 subjects (4 with vomiting) • GI AEs in the LD regimens were minimal ➢ 1100/550 mg = 1 subject with mild nausea ➢ 1650/825 mg = None • Transient low-level bilirubin increases were seen at 1650 mg MD and both LDs consistent with Mrp2 & Bsep inhibition • No clinically significant ECG or PE changes • CEM-102 was well tolerated at MDs up to 1100 mg and in both LD regimens

Pharmacokinetics

Descriptive statistics of CEM-102 plasma PK parameters are summarized in the following tables & figures.

Table 1. Mean PK Parameters for CEM-102 in Plasma after Single and Multiple Doses

Parameter	Cohort 1 550 mg Mean	Cohort 1 550 mg SD	Cohort 2 1100 mg Mean	Cohort 2 1100 mg SD	Cohort 3 1650 mg Mean	Cohort 3 1650 mg SD	Cohort 4 2200 mg Mean	Cohort 4 2200 mg SD
Period 1 Day 1								
C_{max}, µg/mL	33.4	12.2	72.2	10.8	102	25.8	128	28.3
T_{max}, h[a]	2.00	(2-3)	3.50	(1-4)	3.00	(2-4)	6.00	(3-8)
K_{el}, h^{-1}[b]	0.0564	0.0130	0.0617	0.0120	0.0498	0.0151	0.0500	0.0159
$T_{1/2}$, h[c]	12.3	2.98	11.2	2.17	13.9	4.29	13.9	4.52
$AUC_{(0-24)}$, µg·h/mL	242	102	844	115	1,260	386	1,690	427
$AUC_{(0-inf)}$, µg·h/mL	441	269	1,100	247	1,800	689	2,650	978
CL/F, L/h	1.69	1.00	1.04	0.202	1.07	0.519	0.924	0.434
V_d/F, L	28.4	10.8	16.9	1.20	21.1	4.96	19.2	6.38
Period 2 Day 6								
C_{max}, µg/mL	130	30.5	281	52.5	324	26.8	-	-
T_{max}, h[a]	3.00	(1.5-4)	4.00	(4-8)	4.00	(1.5-6)	-	-
K_{el}, h^{-1}[b]	0.0554	0.0131	0.0404	0.0162	0.0199	0.0118	-	-
$T_{1/2}$, h[c]	12.5	3.05	17.1	6.79	31.6	18.9	-	-
$AUC_{(0-12)}$, µg·h/mL	1,150	433	2,530	417	3290	146	-	-
CL/F, L/h	0.553	0.255	0.449	0.100	0.0503	0.0223	-	-
V_d/F, L	9.88	3.05	12.7	5.66	34.8	22.9	-	-
C_{max} accumulation ratio	3.89	-	3.89	-	3.18	-	-	-
$AUC_{(0-12)}$ accumulation ratio	2.61	-	2.30	-	1.83	-	-	-

a Expressed as median and range
b Apparent first-order terminal elimination rate constant
c Expressed as harmonic mean and pseudo SD

Figure 1. Mean CEM-102 Plasma Concentrations after Single and Multiple Doses

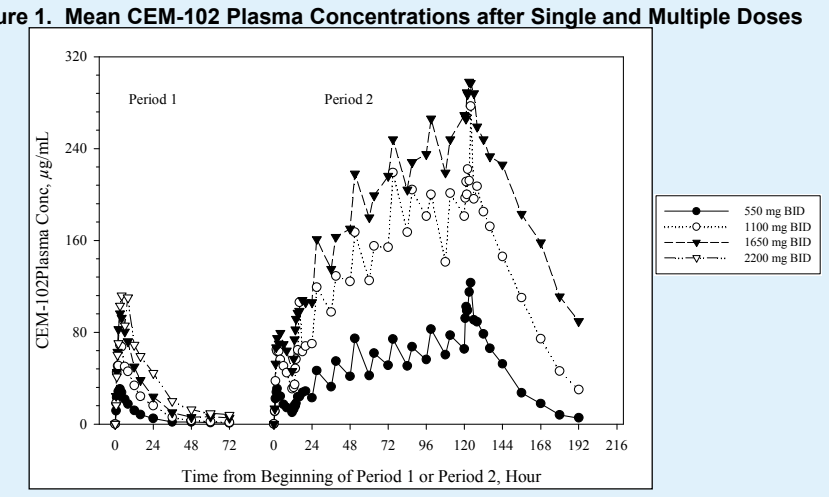

Period 1
Period 2
CEM-102Plasma Conc, µg/mL
Time from Beginning of Period 1 or Period 2, Hour
550 mg BID
1100 mg BID
1650 mg BID
2200 mg BID

Table 2. Mean PK Parameters for CEM-102 in Plasma after Loading Doses on Day 1 Followed by Maintenance Doses X 6.5 Days

Parameter	1100/550 mg Mean	1100/550 mg SD	1650/825 mg Mean	1650/825 mg SD
C_{max}, µg/mL	144	37.9	261	32.4
T_{max}, h[a]	4.00	(3-6)	3.50	(3-4)
K_{el}, h^{-1}[b]	0.0427	0.0139	0.0231	0.00650
$T_{1/2}$, h	16.3	5.60	30.0	8.50
$AUC_{(0-12)}$, µg·h/mL	2,380	717	2,580	345
CL/F, L/h	0.246	0.0618	0.325	0.0397
V_d/F, L	6.04	1.70	15.0	4.86

a Expressed as median and range
b Apparent first-order terminal elimination rate constant

Figure 2. Mean CEM-102 Plasma Concentrations after Single and Multiple Doses

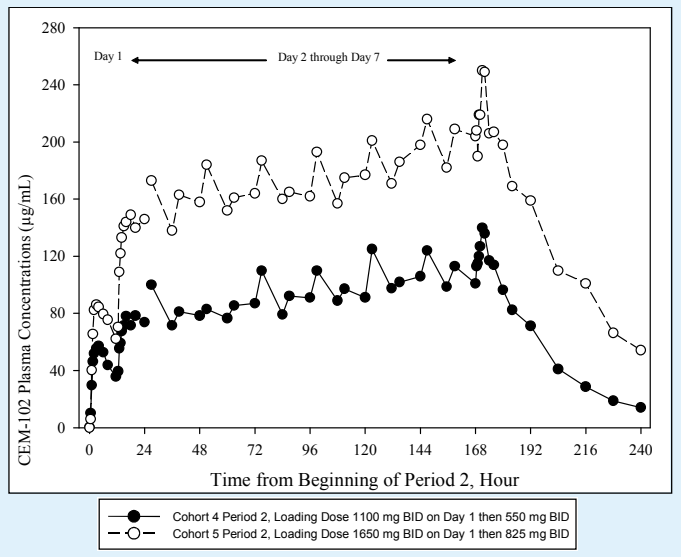

Day 1
Day 2 through Day 7
CEM-102 Plasma Concentrations (µg/mL)
Time from Beginning of Period 2, Hour
Cohort 4 Period 2, Loading Dose 1100 mg BID on Day 1 then 550 mg BID
Cohort 5 Period 2, Loading Dose 1650 mg BID on Day 1 then 825 mg BID

Conclusions

- C_{max} and AUC showed more than dose proportional increases from 550 mg to 1100 mg, then approximately dose proportional increases from 1100 mg to 2200 mg (single dose only for 2200 mg).
- Accumulation occurred from Day 1 to last day of dosing at all dose levels
- 1100 mg BID Loading Dose followed by 550 mg BID Maintenance Dose resulted in trough plasma concentrations of 74 µg/mL at 24 hours and 101 µg/mL after 7 days of dosing
- 1650 mg BID Loading Dose followed by 825 mg BID Maintenance Dose resulted in trough plasma concentration of 146 µg/mL at 24 hours and 204 µg/mL after 7 days of dosing.
- All dose levels in all regimens were considered safe in healthy subjects.
- Single doses to 1650 mg and multiple doses to 1100 mg were well tolerated by healthy subjects.
- The 1650 and 1100 BID LD regimens were very well tolerated by healthy subjects.
- A LD regimen intermediate between the 2 regimens evaluated (e.g. 1500 mg BID followed by 600 mg BID) should provide the optimal exposure for eradication of both *S. aureus* and Group A β-hemolytic streptococci and minimize any potential for emergence of resistance.