252

Molecular Characterization of Off-Target Activities of Telithromycin, a Macrolide/Ketolide Antibiotic:The Role of Nicotinic Acetylcholine Receptors

D. Bertrand[1], S. Bertrand[1], E. Neveu[1], P. Fernandes[2]

[1]HiQScreen Sàrl, Geneva, Switzerland, [2]Cempra Pharmaceuticals, Chapel Hill, NC

HiQScreen Sàrl
15, rue de l'Athénée
1206 Geneva
Switzerland
+41 786964656

Abstract

Macrolide antibiotics are selective bacterial protein synthesis inhibitors. Clinical usage of this class over five decades has resulted in emergence of resistance. Teltithromycin (Ketek™) is a recently described macrolide of the ketolide subclass containing a pyridine moiety. It has improved antimicrobial activity, but is associated with adverse clinical effects including visual dysfunction, exacerbation of myasthenia gravis symptoms, and liver failure, collectively called the "Ketek Effects". The repercussions from the approval of telithromycin have adversely affected the development of new macrolides as a class and new antibiotics on the whole. Future developments of macrolides require a delineation of the structure - toxicity relationships underlying these adverse effects

Introduction

- Macrolide antibiotics are potent antibiotics that have enjoyed decades of use. Resistance to older macrolides is increasing and in pneumococci it is approximately 35% (ref). Telithromycin (Ketek™) was developed to meet the need for a new macrolide antibiotic for treating drug resistant pathogens but it use became limited because of unexplainable side effects were observed including aggravation of Myasthenia Gravis, blurred vision, and liver toxicity (collectively called the "Ketek effects").
- Characterization of the reason for these off-target activities is important to the development of future macrolides/ketolides.
- Aggravation of the Myasthenia Gravis symptoms and reversible visual disturbances suggests a possible interaction of macrolides with neuronal nicotinic acetylcholine receptors. As these receptors form a large family of ligand-gated channels that are widely expressed, we determined how macrolides/telithromycin could potentially interfere.

Materials and Methods

Oocytes preparation and injection

- *Xenopus* oocytes were prepared and injected with cDNAs encoding for the human $\alpha3\beta2$, $\alpha3\beta4$, $\alpha4\beta2$, $\alpha7$ and $\alpha1\beta1\delta\varepsilon$ (NMJ). At least one hundred oocytes were injected using a proprietary automated injection device and receptor expression was examined two or more days later.
- A small piece of ovary was isolated for immediate preparation while the remaining part was placed at 4ºC in a sterile Barth solution containing in mM NaCl 88, KCl 1, $NaHCO_3$ 2.4, HEPES 10, $MgSO_4.7H_2O$ 0.82, $Ca(NO_3)_2.4H_2O$ 0.33, $CaCl_2.6H_2O$ 0.41, at pH 7.4, and supplemented with 20 µg/ml of kanamycin, 100 unit/ml penicillin and 100 µg/ml streptomycin. All recordings were performed at 18ºC and cells superfused with OR2 medium containing in mM : NaCl 82.5, KCl 2.5, HEPES 5, $CaCl_2.2H_2O$ 1.8, $MgCl_2.6H_2O$ 1, pH 7.4, and 1 µM atropine was added to prevent possible activation of endogenous muscarinic receptors.

Electrophysiological recordings

- Currents evoked by ACh or other agonists were recorded using an automated process equipped with standard two-electrode voltage-clamp configuration (TVEC). Unless indicated, cells were held at –70 mV. Data were captured and analyzed using a HiQScreen proprietary data acquisition and analysis software running under Matlab (Mathworks Inc.).

Data Analysis and Statistics

- For statistical analysis, values were computed either with Excel (Microsoft) or Matlab (Mathworks Inc.). All experiments were carried out using at least three to five cells. Only cells displaying a stable holding current and robust ACh-evoked current measured in control conditions were considered for the analysis.

Results and Discussion

- Effects of macrolides were examined at four subtypes of nicotinic acetylcholine receptors (NMJ, $\alpha3\beta4$, $\alpha4\beta2$ and $\alpha7$). While NMJ receptors are exclusively expressed on the muscle receptor membrane, the $\alpha3\beta4$ receptors are preponderantly expressed in cholinergic ganglia. The distribution of the receptors are shown in **Figure 1**.
- None of the macrolides evoked a current in nACh receptor expressing cells indicating that these compounds do not act as agonists. A dose –dependent *reduction* of the responses evoked by ACh was observed upon addition of a low concentration of telithromycin (2 µM) (**Figure 2)**.
- The relative activities of four macrolides, clarithromycin, azithromycin, telithromycin and the novel fluoroketolide, solithromycin (CEM-101), were tested at the four nicotinic acetylcholine receptors (**Figure 3**). Measurements were made after sustained exposure (20 minutes) and recording the acetylcholine evoked currents measured at periodic intervals. This protocol is thought to mimic the conditions experienced in vivo. The fraction of acetylcholine-evoked current measured at the end of the twenty minutes incubation versus the amplitude of current measured in control were used for quantification.
- The $\alpha3\beta4$ and the $\alpha7$ receptors found in the ciliary ganglion of the eye are responsible for regulating visual accommodation. Telithromycin inhibited both these receptors $\geq$ 90%, whereas lesser inhibition was observed with clarithromycin, azithromycin and solithromycin. This effect is enhanced by the presence of progesterone, explaining why the visual effects are more common in younger women administered telithromycin.
- Comparable degree of inhibition at the neuromuscular junction were obtained with all macrolides suggesting that this mechanism alone cannot explain the risk of muscle failure in myasthenic patients reported for telithromycin. The putative telithromycin metabolites, pyridine-imidazole and pyridine-imidazole-n-oxide markedly inhibited the neuromuscular junction receptors (**Figure 4**). and also inhibited up to 25 % the $\alpha3\beta2$ nAChR, expressed in the presynaptic ending of the neuromuscular junction. Therefore, telithromycin and its metabolites inhibit both pre- and postsynaptic receptors.
- The $\alpha7$ nAChRs is known to regulate inflammation and is found at the ends of the Vagus nerve innervating the liver. Stimulation of the $\alpha7$ nAChRs protects against Fas-apotosis in the mouse liver. On the contrary, vagotomized animals showed exacerbation of liver Fas-apoptosis confirming the role of cholinergic neurotransmission. Telithromycin, but not solithromycin and older macrolides (**Figure 4**), inhibits the $\alpha7$ receptors which are critical to protecting the liver from inflammation.
- Concentration inhibition curves were determined (**Figure 5**). Telithromycin preferentially inhibited $\alpha3\beta4$ receptors with an IC_{50} of 0.1 µM. At 10 µM telithromycin almost abolished the ACh-evoked current at this receptor subtype. At least a ten-fold shift toward lower sensitivity was observed with azithromycin and clarithromycin. Data obtained with CEM-101, which shares the macrolide ring of telithromycin, *but not the pyridine moiety*, show a clear difference in the inhibition profile (**Figure 6**). A similar trend was seen with the $\alpha7$ receptor with telithromycin causing the largest inhibition and an IC_{50} of about 0.15 µM. It is noteworthy to observe that inhibition reaches a plateau at 30% for clarithromycin. Even at the highest concentration of clarithromycin, about 60% of the ACh response remains whereas only 10% or less of the response would be observed for telithromycin. Comparison of telithromycin and CEM-101 inhibition further underlines the relevance of the pyridine moiety emphasized by the difference in IC_{50}'s between these two ketolides. At the $\alpha4\beta2$ nAChRs, which are the high nicotine affinity receptors in the central nervous system (CNS), concentration inhibition curves also show the highest sensitivity to telithromycin; the IC_{50} was about 4 µM which is significantly higher than that observed for $\alpha3\beta4$ or $\alpha7$ nAChRs. Moreover, azithromycin and clarithromycin displayed even higher IC_{50}'s.

Figure 1 Nicotinic ACh receptor subtypes

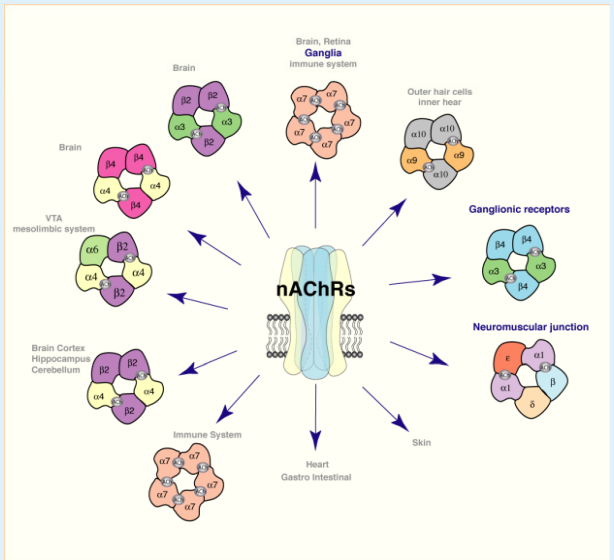
nAChRs
Brain, Retina
Ganglia
Immune system
Outer hair cells
Inner hear
Ganglionic receptors
Neuromuscular junction
Skin
Heart
Gastro Intestinal
Immune System
Brain Cortex
Hippocampus
Cerebellum
VTA
mesolimbic system
Brain

Figure 2

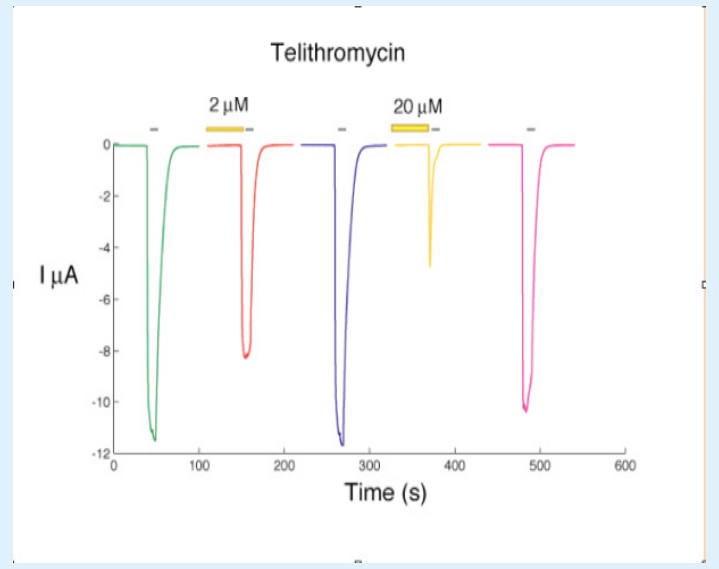
Telithromycin
2 µM
20 µM
I µA
Time (s)

Figure 3

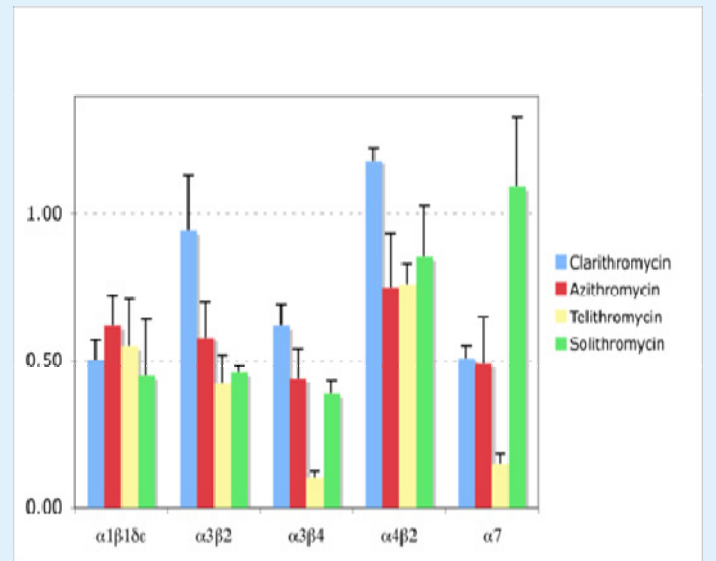
Clarithromycin
Azithromycin
Telithromycin
Solithromycin
1.00
0.50
0.00
α1β1δε
α3β2
α3β4
α4β2
α7

Figure 4 Effects of three putative metabolites of telithromycin

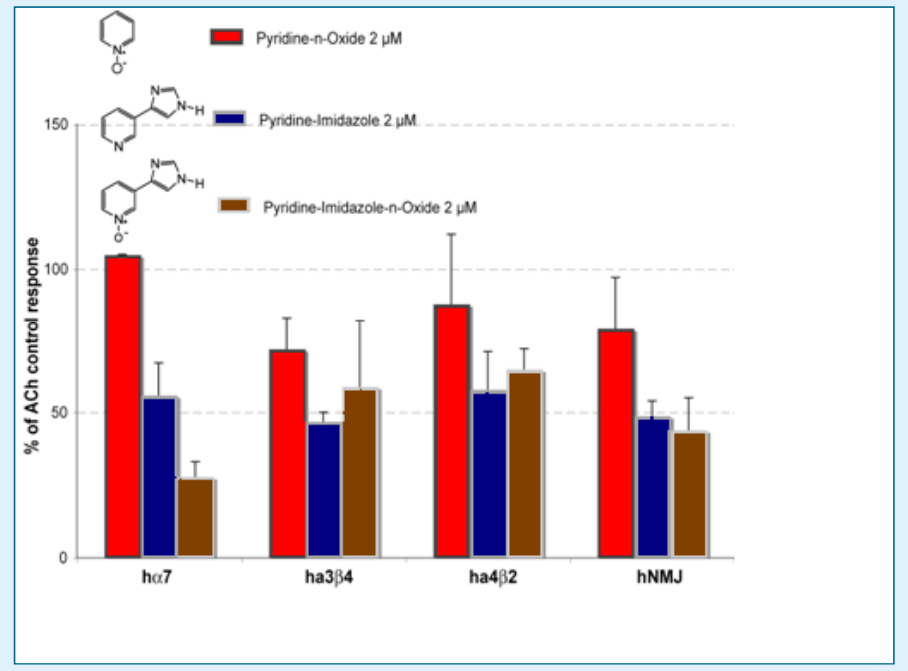
Pyridine-n-Oxide 2 µM
Pyridine-Imidazole 2 µM
Pyridine-Imidazole-n-Oxide 2 µM
% of ACh control response
hα7
ha3β4
ha4β2
hNMJ

Plot of the maximum inhibition observed for each of the metabolites for four nAChRs subtypes clearly illustrate the differential effects observed at the receptors. Note the largest inhibition observed at $\alpha7$ with pyridine-imidazole-n-oxide.

Figure 5 Inhibition of ganglionic and central nAChRs by four macrolides

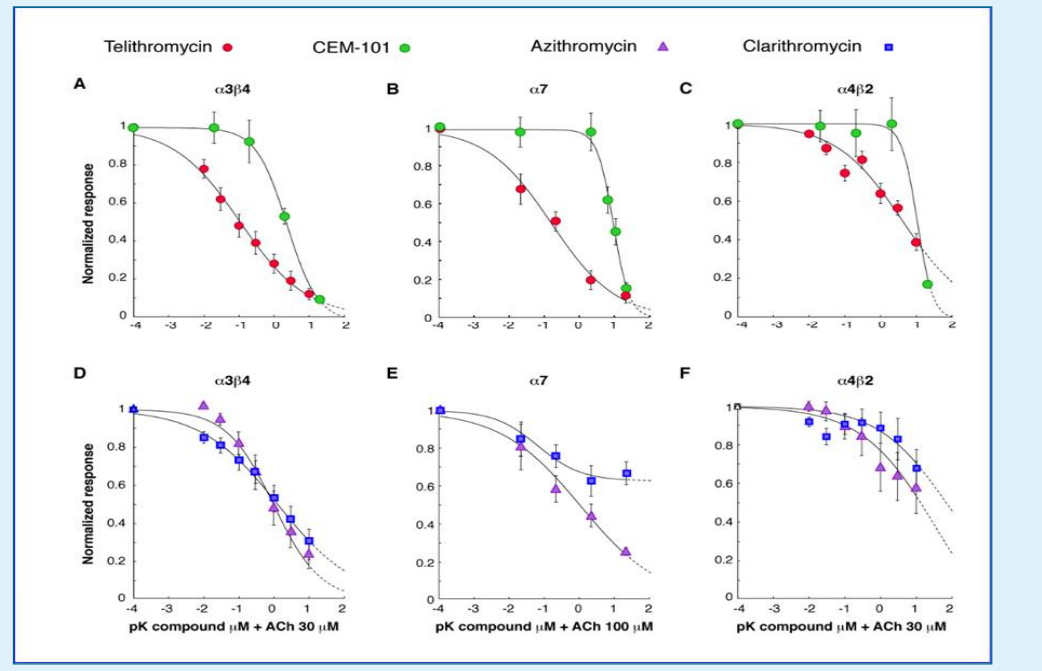
Telithromycin
CEM-101
Azithromycin
Clarithromycin
Normalized response
pK compound µM + ACh 30 µM
pK compound µM + ACh 100 µM

A-C) Concentration inhibition curves of $\alpha3\beta4$, $\alpha7$ and $\alpha4\beta2$ caused by telithromycin (filled circles) and the novel ketolide CEM-101 (open circles). **D-F**) Concentration inhibition curves of $\alpha3\beta4$, $\alpha7$ and $\alpha4\beta2$ caused by azithromycin (open triangles) and the clarithromycin (open squares).

Figure 6

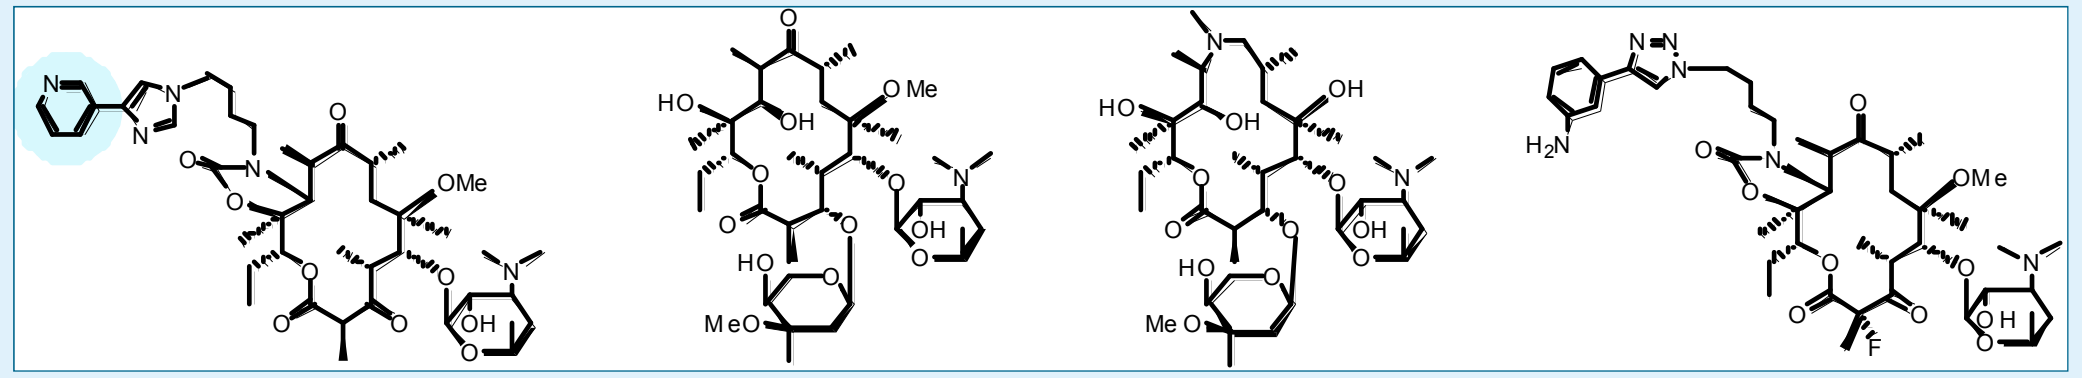

Telihromycin | Clarithromycin | Azithromycin | **CEM-101**

Chemical structure of the macrolides tested at the human nAChRs. The pyridine moiety of telithromycin has been highlighted for clarity

Conclusions

- Inhibition of multiple nACh receptors, at various locations in the body, may explain the off-target effects of telithromycin.
- The pyridine moiety of telithromycin could be the link to the adverse events associated with telithromycin and the "Ketek effects" could simply be "pyridine effects".
- These results should be useful for guiding future macrolide/ketolide development.

References

1. Omura *Macrolide Antibiotics Chemistry, Biology and Practice* (2002).
2. Dani JA, Bertrand D. Nicotinic acetylcholine receptors and nicotinic cholinergic mechanisms of the central nervous system. *Annu Rev Pharmacol Toxicol* **47,** 699-729 (2007).
3. Berg DK, Conroy WG. Nicotinic alpha 7 receptors: synaptic options and downstream signaling in neurons. *J Neurobiol* **53,** 512-523 (2002).
4. Sargent PB. Nicotinic receptors concentrated in the subsynaptic membrane do not contribute significantly to synaptic currents at an embryonic synapse in the chicken ciliary ganglion. *J Neurosci* **29,** 3749-3759 (2009).
5. Borovikova LV, Ivanova S, Zhang M, *et al.* Vagus nerve stimulation attenuates the systemic inflammatory response to endotoxin. *Nature* **405**, 458-462 (2000).
6. Tsuneki H, Kimura I, Dezaki K, *et al.* Immunohistochemical localization of neuronal nicotinic receptor subtypes at the pre- and postjunctional sites in mouse diaphragm muscle. *Neurosci Lett* **196**, 13-16 (1995).
7. Hiramoto T, Chida Y, Sonoda J, *et al*. The hepatic vagus nerve attenuates Fas-induced apoptosis in the mouse liver via alpha7 nicotinic acetylcholine receptor. *Gastroenterology* **134**, 2122-2131 (2008).